HEALING OUTCOMES OF STALLED WOUNDS USING A NOVEL TOPICAL ANTIMICROBIAL SUSPENSION

Tammy Cauthorne-Burnette, MSN, FNP-BC, WCC; Deena Conrad-Vlasak, MS, CCRA; Brad Burnam, MA, MS

Introduction

The care and management of patients with chronic, non-healing wounds is an expensive, time-consuming, and challenging task. Many therapies are often tried without success on non-responsive wounds with the goal of finding a treatment to trigger the healing process. Overcoming the factors that contribute to delayed healing are the key components and primary challenge to the treatment of chronic wounds. (1) The novel antimicrobial gel CurX™ proprietarily combines antimicrobial agents in a petroleum based therapy for the topical treatment of wounds. This gel has been employed to treat a series of wounds with varying etiologies of wound development. The purpose of these case studies was to evaluate the hypothesis that the topical application of a novel antimicrobial gel would initiate healing in patients with a history of poor and/or delayed wound healing.

Methods

The focus of this series of case studies is topical wound care. Three case studies are presented in which patients were chosen due to stalled wound healing or a history of poor wound healing. A thin layer of CurX™ antimicrobial gel was applied to and around the affected area. The area was covered and dressings changed according to facility protocol.

Results

The use of the novel antimicrobial suspension, CurX™, successfully initiated healing in the wounds presented. This rapid transformation in healing resulted in increased patient and clinician satisfaction, as well as an enhanced cost savings. While not anticipated, a dressing with the CurX ™ left in place for one month remained intact with no occurrence of adverse effects; the wound remained stable and ulcer-free.

Conclusions

Based on the outcomes from these case studies, further research is encouraged to determine the effect of CurX™ on the rate and degree of healing in wounds with poor and delayed healing.

Case Study 1
Foot Ulcers

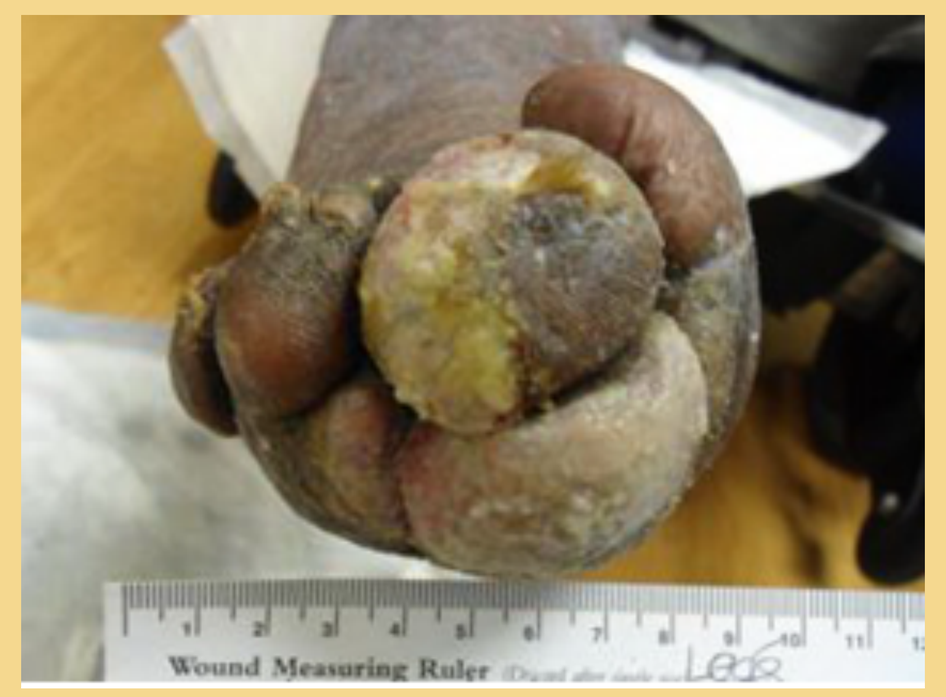
Baseline

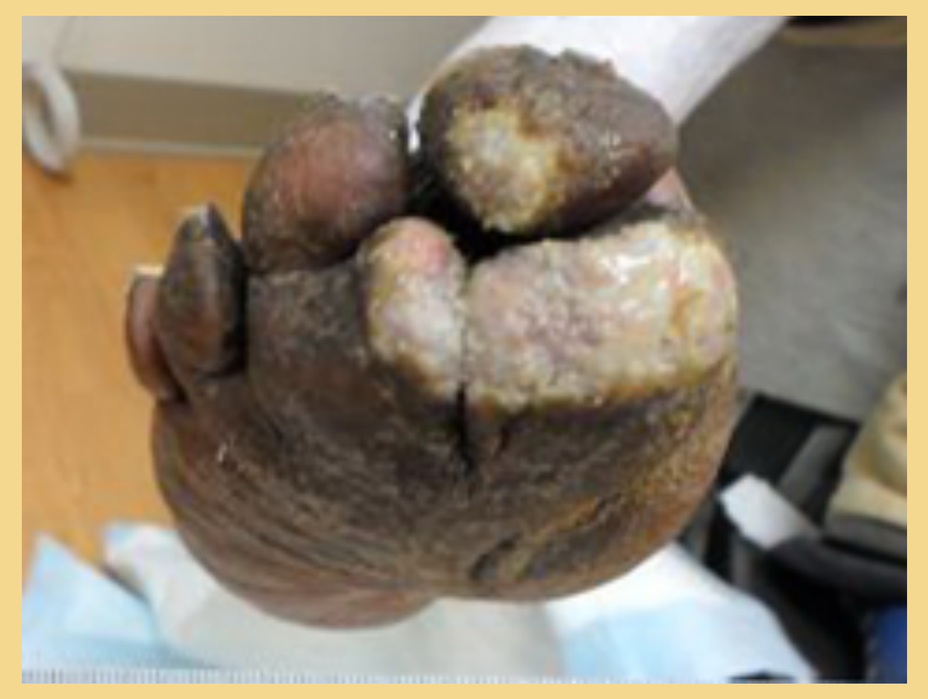
1 Month

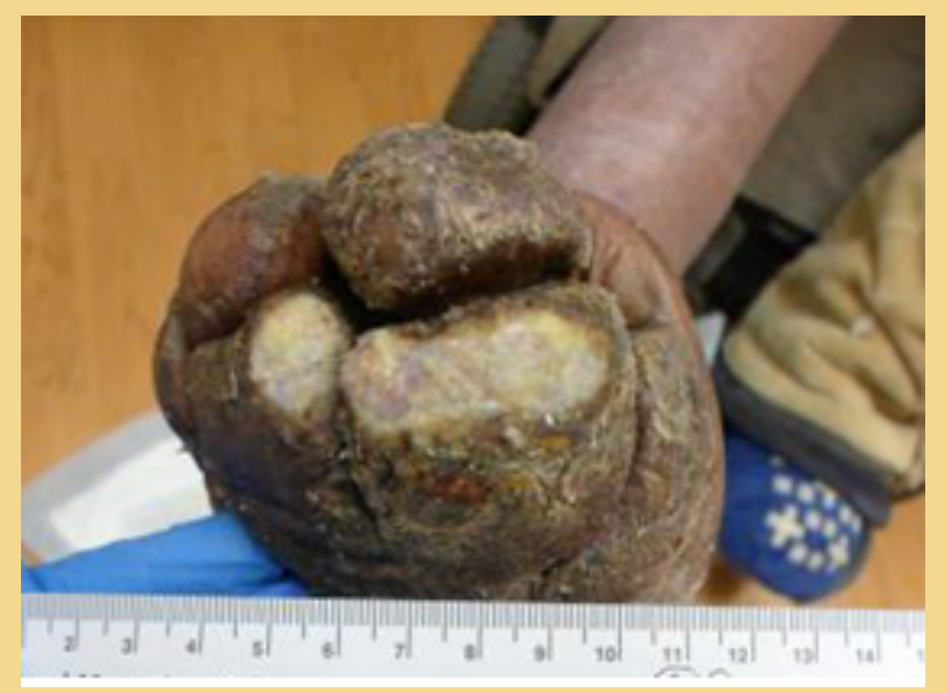
Month 2

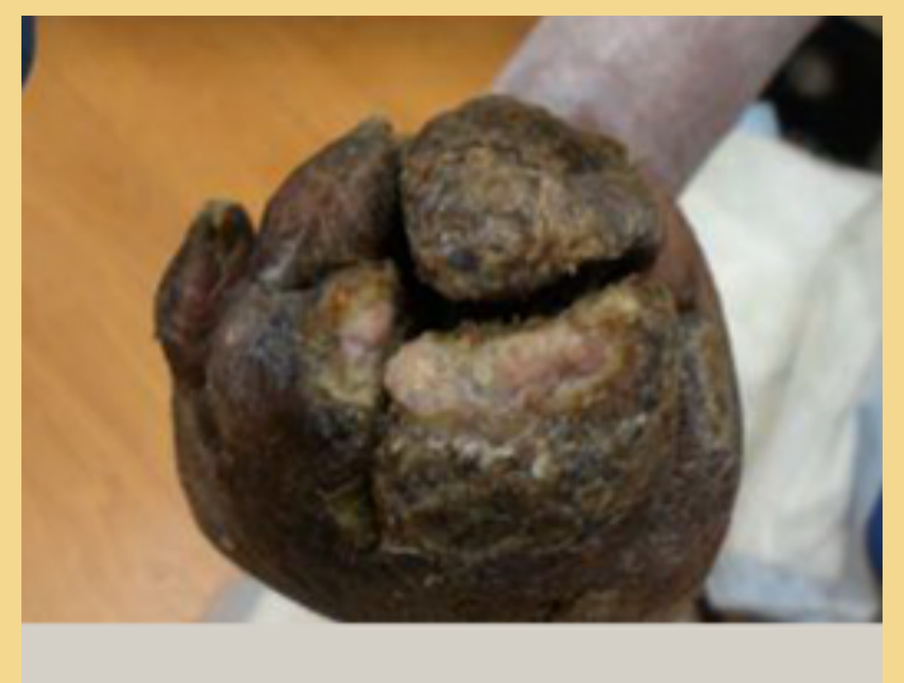
Month 3

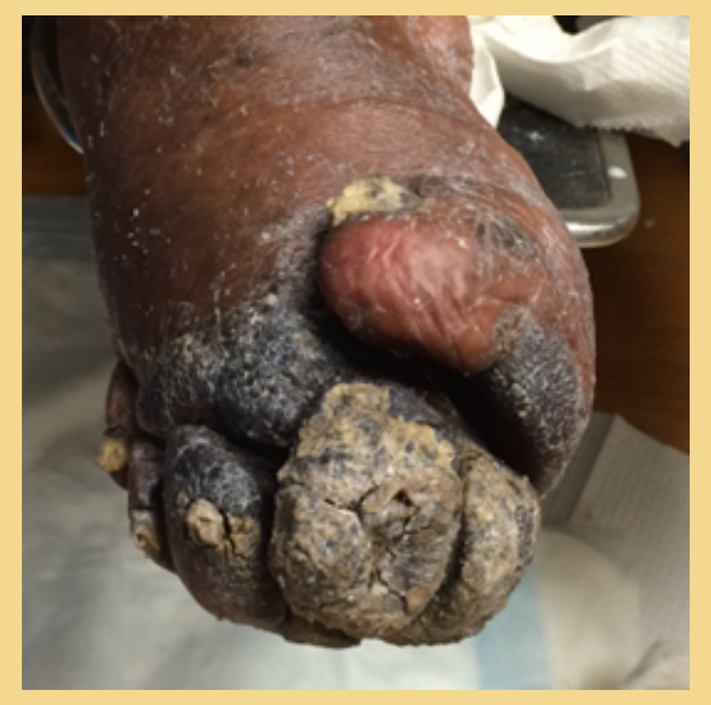
One month without a dressing change.

Patient Outcome: After approximately 3 months of treatment the wounds were dry and healed. No ulceration occurred with a dressing left on for > 30 days

Case Discussion: A 56 year old with a medical history of chronic lymphedema, DM, HTM, Hyperlipidemis, CVA with residual hemiparesis of R side, wheelchair bound and tobacco dependant was seen in the clinic for developing venous foot ulcers. Original treatment consisted of butadiene gauze wraps and isosorb with kling, ace and Coban wraps. Wound care was delivered twice weekly for 7 weeks with little improvement. Patient was then given Clindamycin again with minimal improvement noted. A trial of CurX™ was initiated on the ulcerations. The patient was followed for 5 months during which time rapid improvement in the wound was observed. After approximately 4 months from initial treatment the patient lost Medicaid, and the last dressing consisting of CurX and gauze with a Coban wrap remained on for over 1 month with no development of ulcerations.

Case Study 2
Lower Extremity Laceration

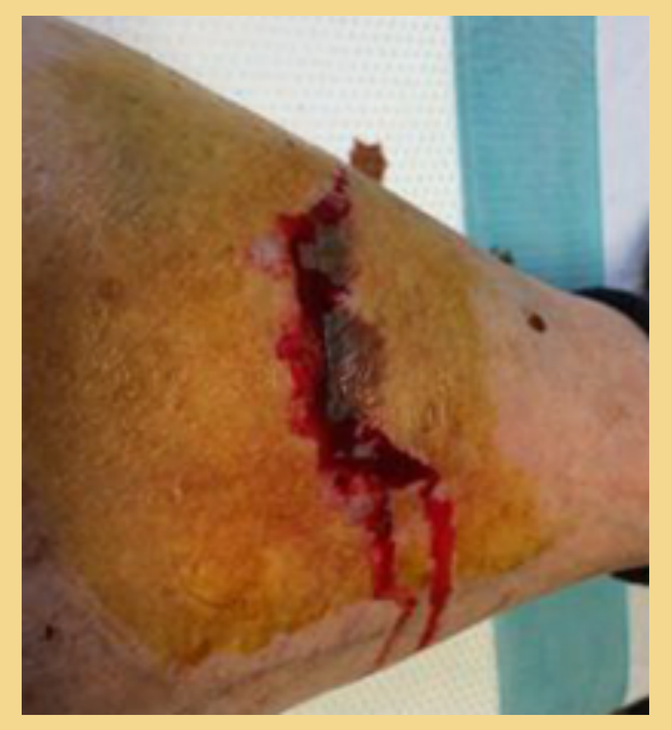
Initial Injury

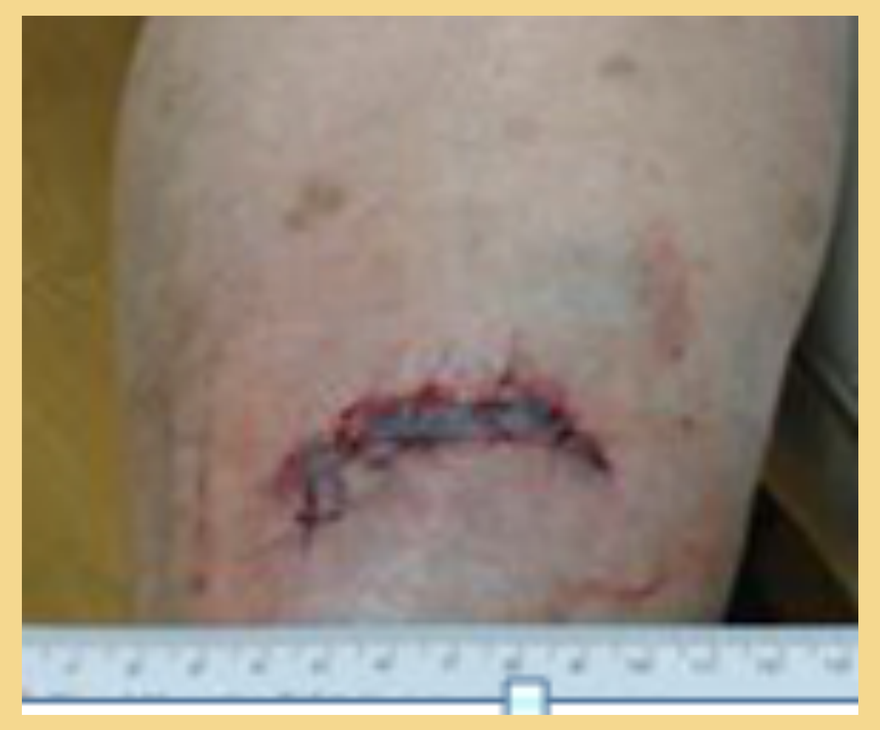
Day 1

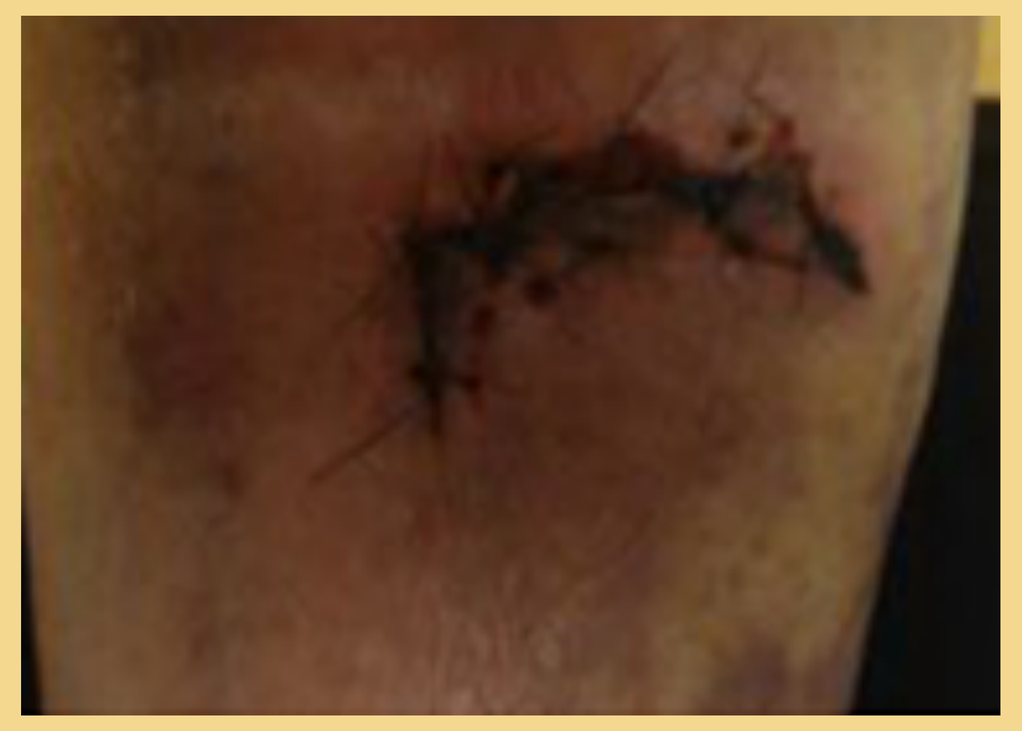
1 Week

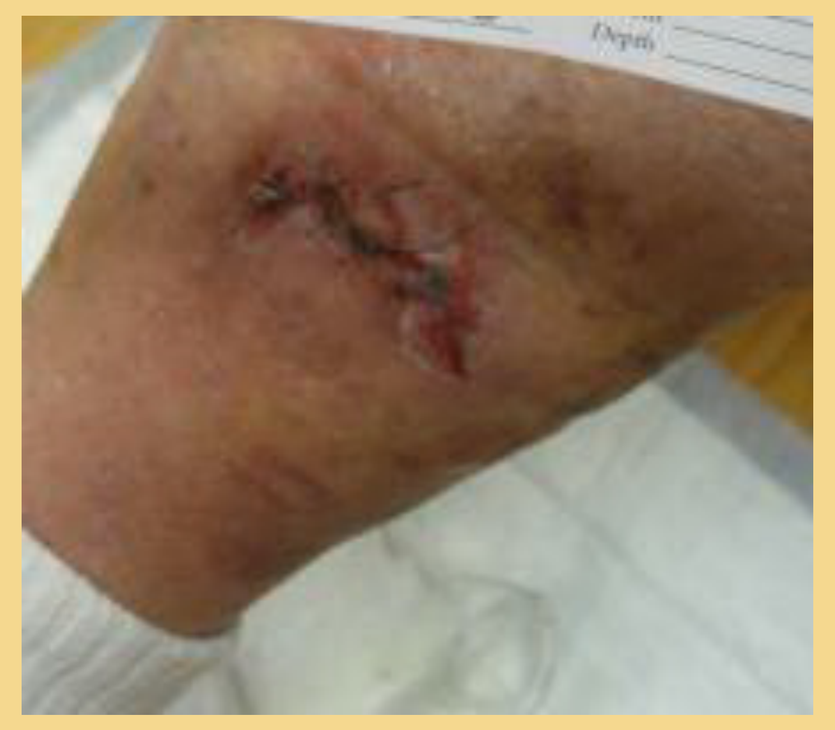
Week 2

Patient Outcome: After approximately 2 weeks, the wound area was closed and healing.

Case Discussion: A 76 year old who was obese, DM insulin dependent, had CKD-4, ASCVD, CABG-4 vessel in 2015 and a history of poor and delayed wound healing presented to clinic 24 hours after laceration to R shin. Based on the patient's history of slow healing, a trial of CurX™ was initiated. The patient was followed for approximately two weeks at which time rapid, improved healing was noted.

Case Study 3
Lower Extremity Mixed Ulcer

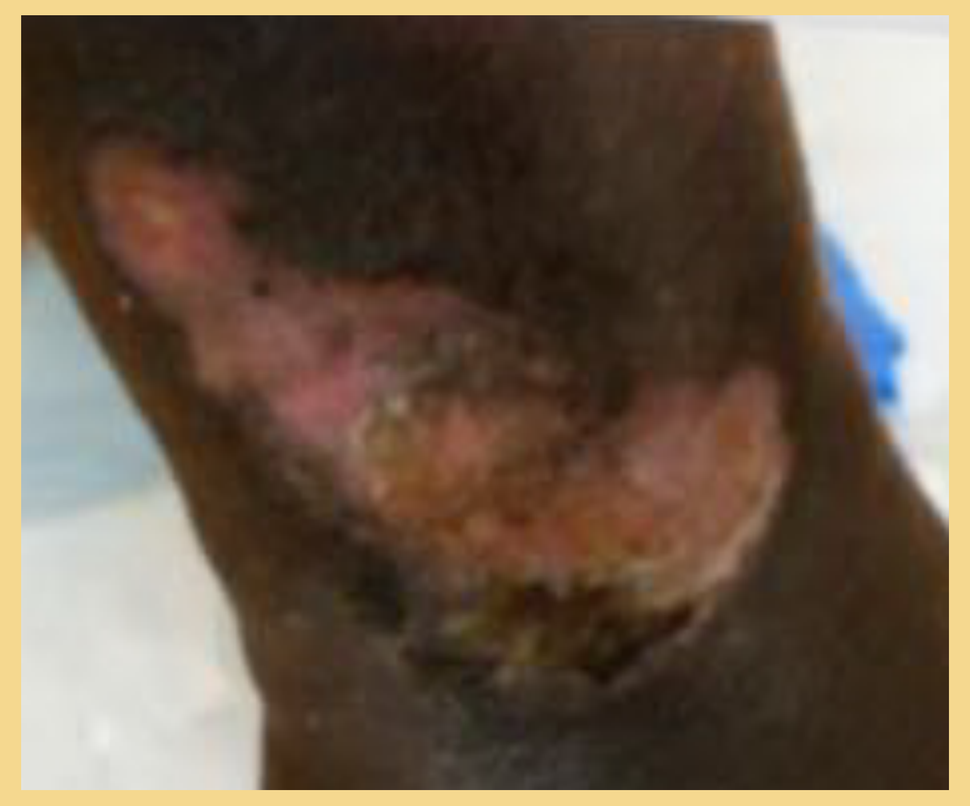
Baseline

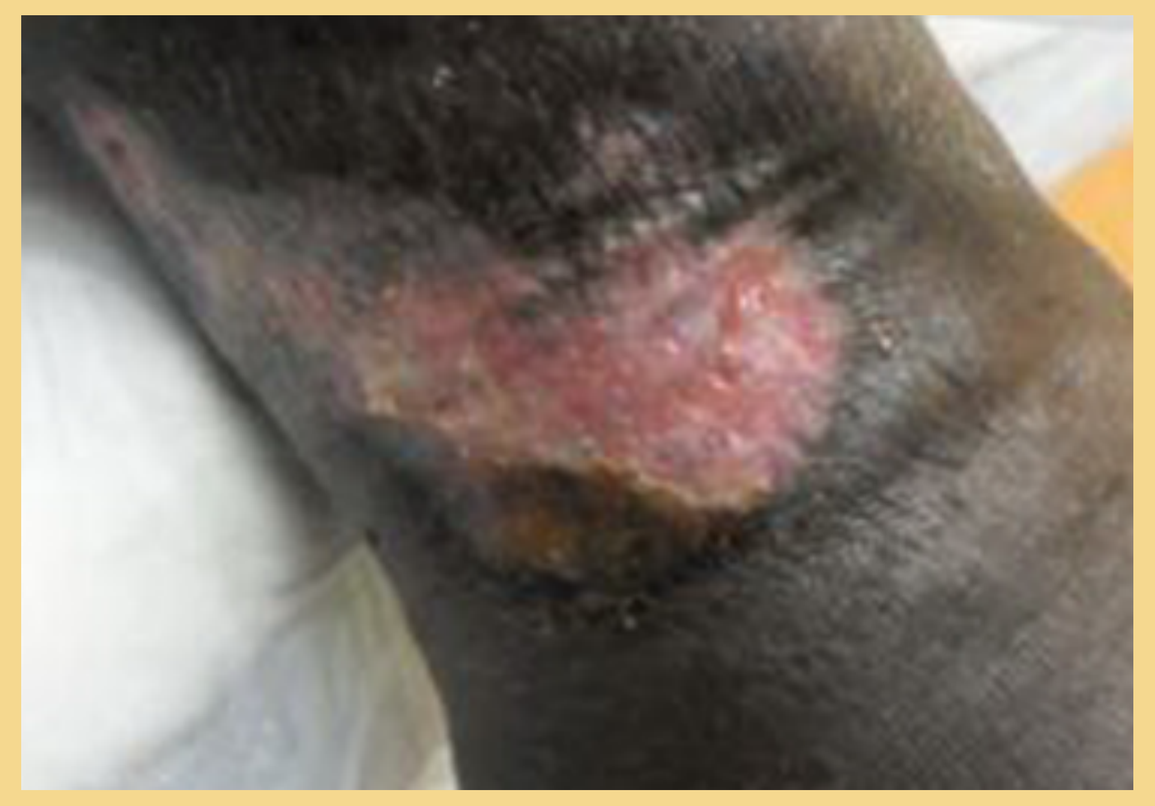
Week 1

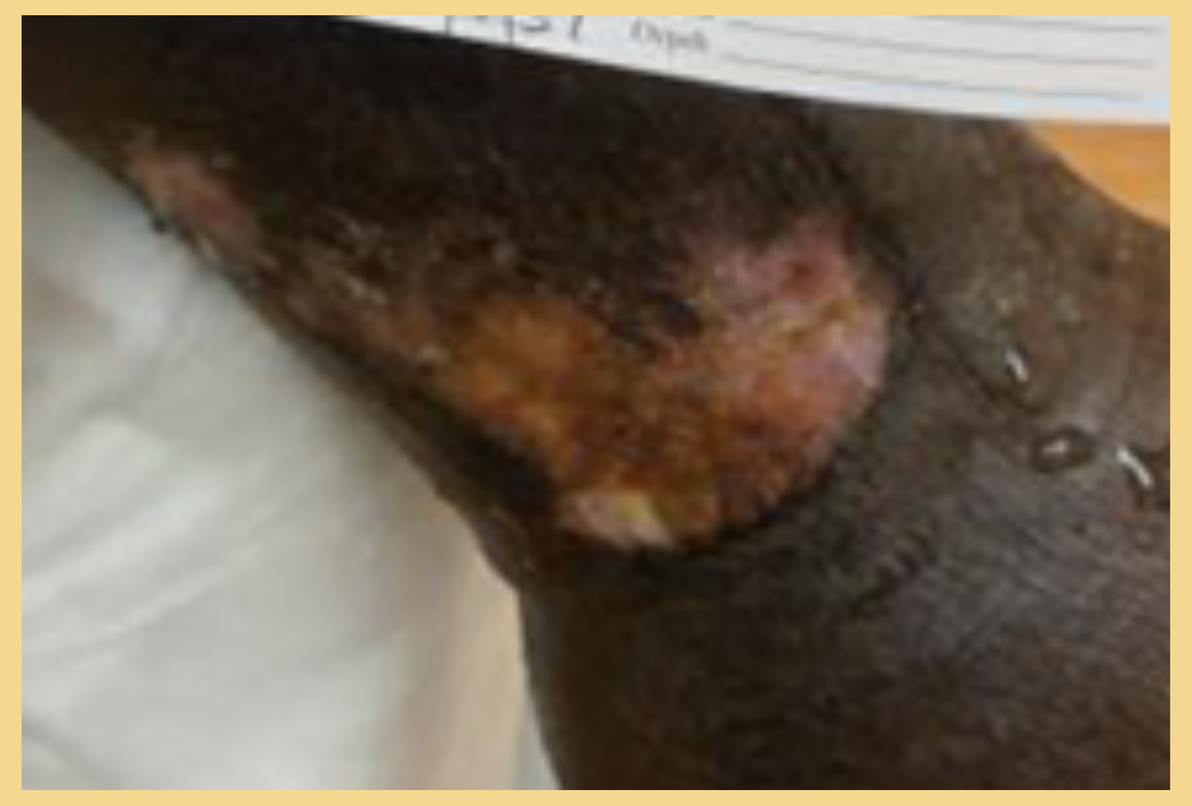
Week 2

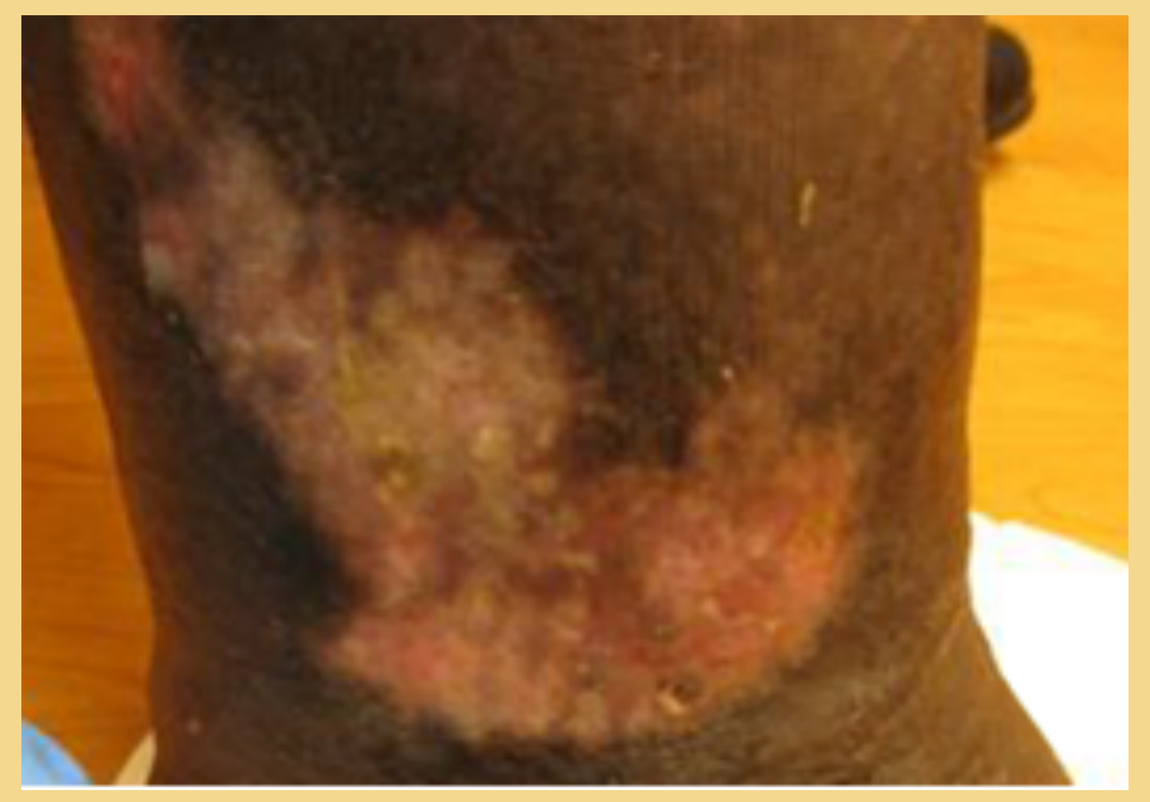
Week 3

Patient Outcome: After 3 weeks of treatment, the wound bed stabilized and re-epithelialization was beginning. Patient was discharged out of the wound clinic 2 weeks ahead of predicted schedule.

Case Discussion: A 76 year-old patient with a history of DM insulin dependency, PAD showing moderate obstruction, diachf, htn, PVD, hyperlipidemia, hypothyroidism, and recurrent mixed ulcers presented with a mixed arterial and venous ulcer of the R lateral lower leg. Initial treatment was twice weekly and consisted of Silvasorb® or Adaptec and gauze and kling. After 6 weeks with minimal improvement, a trial of CurX™ was initiated. The patient was followed for 10 weeks at which time wound stabilization was achieved and re-epithelialization was beginning. A reduction in patient discomfort during dressing changes was observed.

(1) R. Fryberg; J. Banks, *Challenges in the Treatment of Chronic Wounds,* Adv Wound Care (New Rochelle). 2015 Sep 1; 4(9): 560–582.